MicroCT Evaluation of a Long-Lasting Blood Pool and Hepatobiliary Contrast Agents Following Multiple Intravenous Injections in Normal Mouse Models

GN Ton[1], WC Dow[2], DA Bakan[2], M Melchior[2], JP Weichert[1]

[1]University of Wisconsin-Madison,WI and [2]Alerion Biomedical Inc., San Diego, CA

Introduction

Exploiting naturally-occurring endogenous lipid metabolism pathways in the body, we have developed a hepatocyte-selective CT contrast agent consisting of a polyiodinated triglyceride (ITG) embedded in a chylomicron-remnant like delivery vehicle. This lipomimetic vehicle thus selectively shuttles the ITG contrast agent lipids it contains to hepatocytes in the liver (Fig 1).

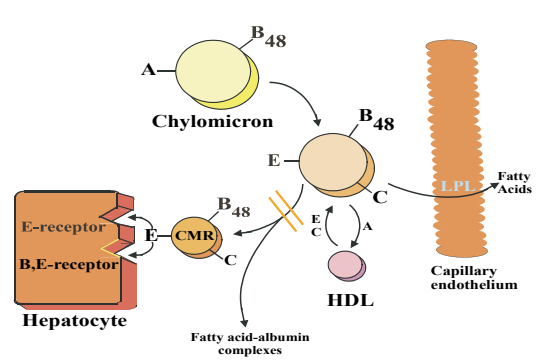
B48
A
Chylomicron
B48
E
C
LPL
Fatty Acids
B48
E-receptor
E
CMR
C
B,E-receptor
E
C
A
HDL
Capillary endothelium
Hepatocyte
Fatty acid-albumin complexes

Fig 1. Hepatic uptake of Chylomicron Remnants

The blood pool version of ITG was developed by incorporating PEG moieties into the phospholipid monolayer shell, resulting in delayed uptake by liver cells and prolonged vascular residence time. Both of contrast agents have demonstrated their ability to provide excellent vascular and hepatobiliary contrast enhancement in both normal and diseased animal models following in a single iv injection [1-4].

Both the liver and blood pool contrast agents have recently become commercially available and known as Fenestra™ LC and VC, respectively. Schematic structures of Fenestra products are shown in Fig 2. In this study we evaluated the in vivo microCT imaging efficacy of Fenestra LC and VC following multiple iv administrations. In addition injection tolerance profiles of the contrast agents were examined.

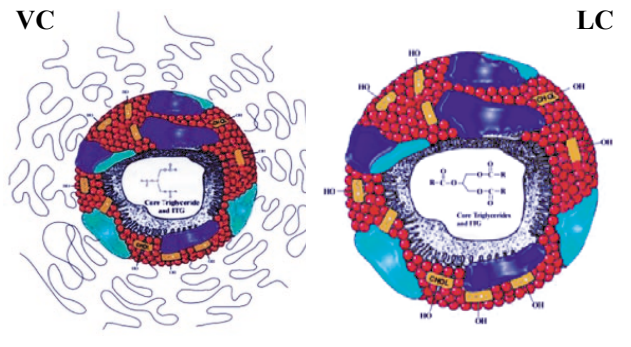
VC
LC

Fig 2. Schematic Structure of Fenestra VC & LC

Materials and Methods

Fenestra LC and VC were obtained from ART Advanced Research Technologies (Montreal, Canada).The final iodine concentration of both formulations was 50 mg I/mL. The contrast agents were administered intravenously to various strains of mice (C57Bl/6, Balb/c, FVB, and SCID) following the dosing schedule shown in Fig 3. Anesthetized animals (n=3) were scanned using a GE eXplore Locus microCT scanner (80 kVp, 450 µA, 200 ms, 400 views and 93 µm³ resolution) or a Siemens microCAT II system (70 kVp, 500 µA, 180 msec, 720 steps and 91 µm³ resolution) prior to and at predetermined time intervals following administration. The animals were placed on a soft diet for at least 24 h prior to initiation of each dosing cycle for minimizing chow-induced abdominal streak artifacts.

Images were visualized and analyzed using both Amira 3-D visualization software (V3.1) and an open-source MicroView program (http://Microview.sf.net). CT values of volumetric ROIs in the inferior vena cava (IVC) and liver were calibrated to Hounsfield Units (HU). Normalized signal intensity (HU/voxel) was obtained from the difference of enhanced SI and the baseline value. The animals were weighed and monitored daily for signs of abnormal behavior for 5 weeks.

Fig 3. Fenestra VC and LC Dosing Regimes in Various Strains of Mice

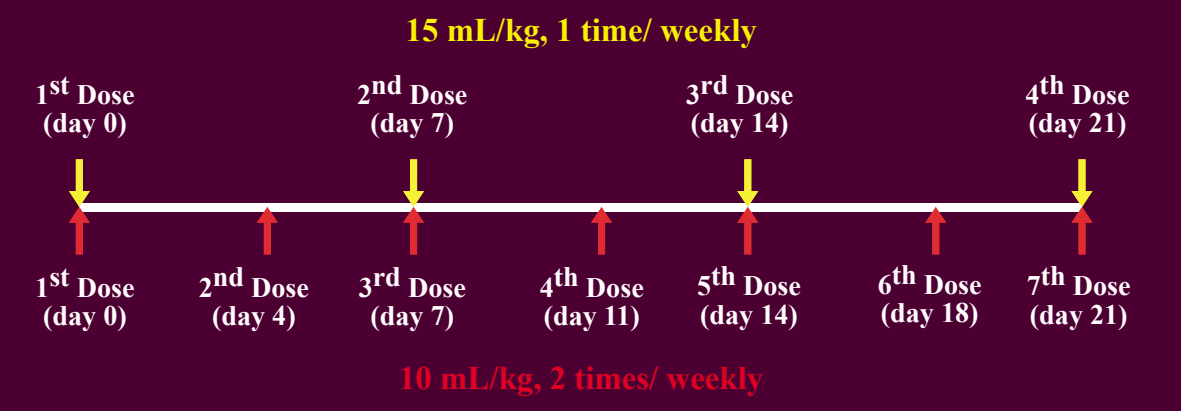
15 mL/kg, 1 time/ weekly
1st Dose (day 0)
2nd Dose (day 7)
3rd Dose (day 14)
4th Dose (day 21)
1st Dose (day 0)
2nd Dose (day 4)
3rd Dose (day 7)
4th Dose (day 11)
5th Dose (day 14)
6th Dose (day 18)
7th Dose (day 21)
10 mL/kg, 2 times/ weekly

Results and Discussion

Fig 4 represents normalized vascular contrast intensities measured in female Balb/c mice at 10 minutes after administration of Fenestra VC (10 or 15 mL/kg) according to the specified dosing regimes (Fig 3). The degree of vascular contrast enhancement at 10 min after each cycle of injections was similar for both 10 and 15 mL/kg dosing regimes.

Fig 4. Normalized vascular contrast intensities of Fenestra VC 10 min following repeating dosing schedules in female Balb/c mice

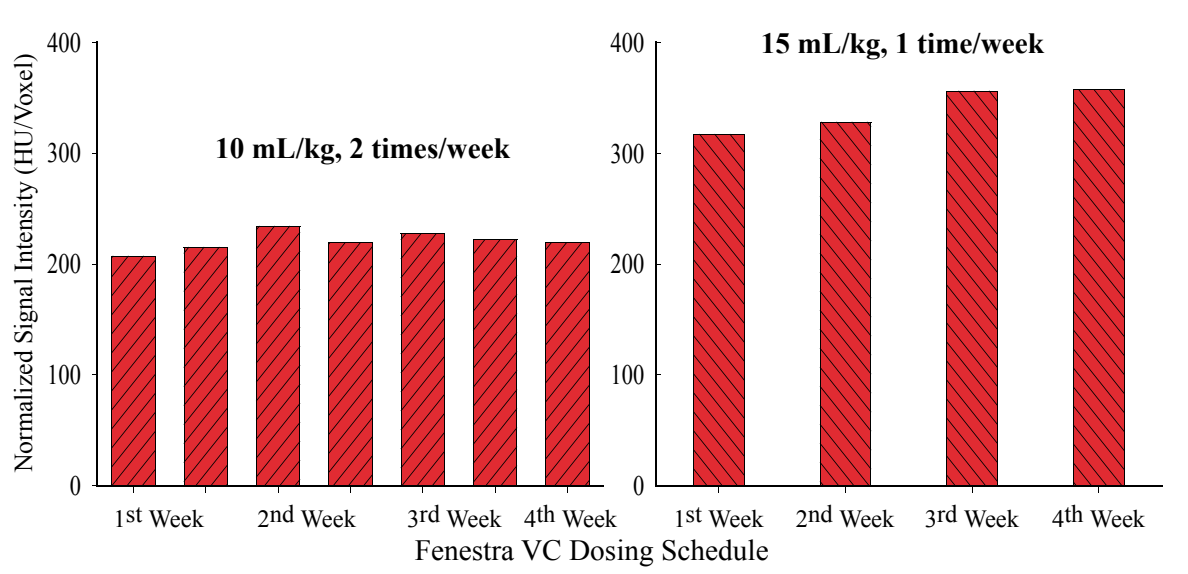
Normalized Signal Intensity (HU/Voxel)
400
300
200
100
0
10 mL/kg, 2 times/week
15 mL/kg, 1 time/week
1st Week
2nd Week
3rd Week
4th Week
1st Week
2nd Week
3rd Week
4th Week
Fenestra VC Dosing Schedule

As shown in Fig 4, normalized vascular contrast intensities achieved after 10 min post injections were directly proportional to the injected doses. FenestraVC was well tolerated in tested mice when given according to the specified dosing schedules. VC-enhanced microCT images of the same Balb/c female mouse received Fenestra VC at a dose of 15 mL/kg weekly for 1 month displayed similar microCT efficacy observed in a single dosing regime, offering flexibility and convenience to perform longitudinal evaluations of the vascular system in the same animal using microCT (Fig 6).

Fig 5. LC-enhanced microCT images of anesthetized C57Bl/6 mice 4 hours following in administration of multiple doses of Fenestra LC (15 mL/kg)

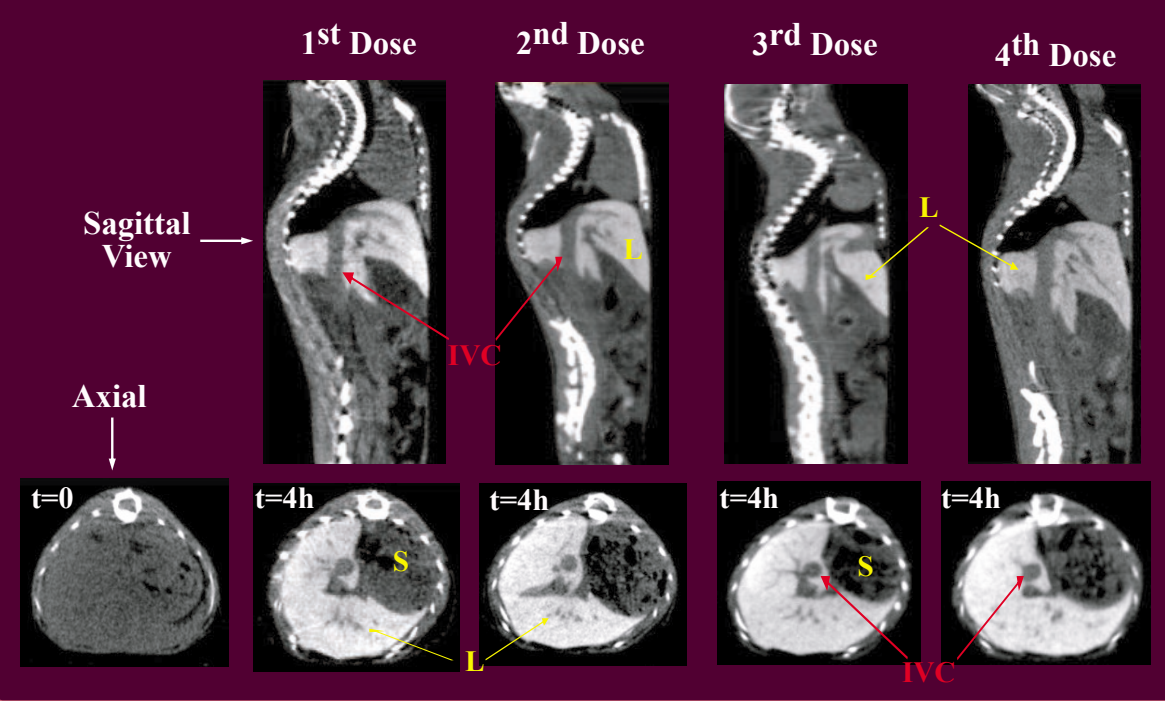
1st Dose
2nd Dose
3rd Dose
4th Dose
Sagittal View
L
L
IVC
Axial
t=0
t=4h
t=4h
t=4h
t=4h
S
S
L
IVC

Excellent liver contrast enhancement was achieved at 4 hours following each cycle of administration of Fenestra LC at 15 mL/kg dose (Fig 5). Non-contrast axial image of an anesthetized C57Bl/6 mouse showed poor soft tissue contrast. It is impossible to accurately delineate the liver. Axial and sagittal microCT images of the mouse obtained after 4 hours following injections displayed excellent liver enhancement, allowing quantitative characterization of the liver to be possible. The IVC and hepatic vascular network became visible after 4 hours due to the negative effect produced by the removal of Fenestra LC from the circulation and its localization in hepatocytes.

Fig 6. VC-enhanced microCT images of an anesthetized Balb/c mouse 4 h following administration of muiltiple doses of Fenestra VC (15 mL/kg)

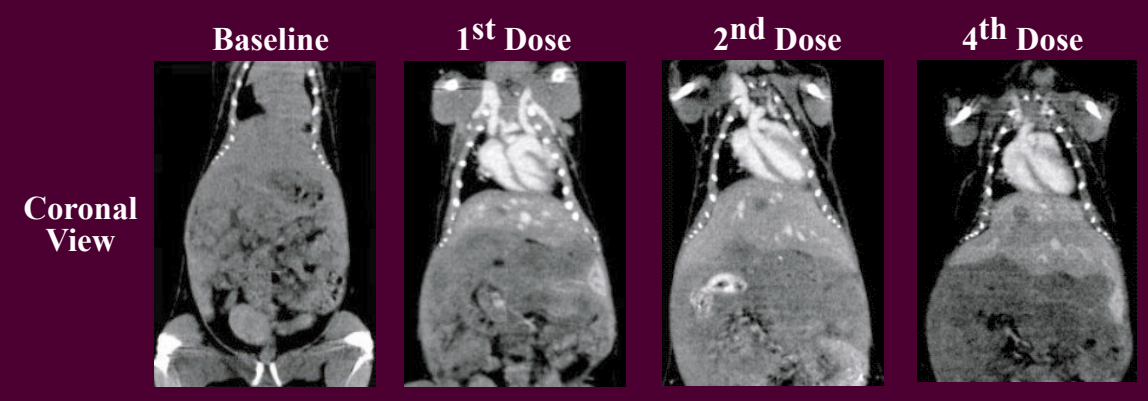
Baseline
1st Dose
2nd Dose
4th Dose
Coronal View

Quantitative analysis of liver signal intensities measured in the same female Balb/c mouse at 4 hours following each weekly dosing cycle is shown in Fig 7. A slightly higher level of liver intensity was observed after 2 rounds of injections at a 15 mL/kg dose, suggesting possible accumulation of the contrast agent or its metabolites in the liver. Fenestra LC was well tolerated in tested mice when given once a week at 15 mL/kg dose for up to 4 weeks.

Fig 7. Normalized liver contrast intensities of Fenestra LC 4 hours following repeating dosing schedules in a female Balb/c mouse

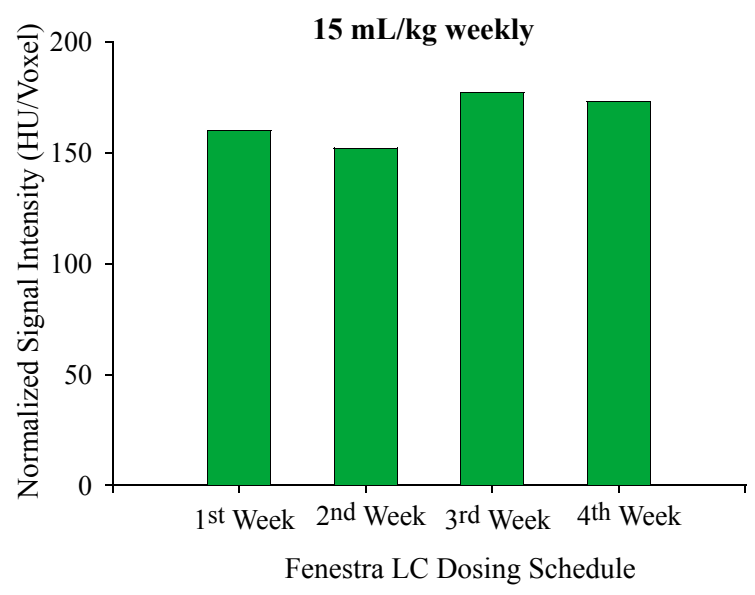
15 mL/kg weekly
Normalized Signal Intensity (HU/Voxel)
200
150
100
50
0
1st Week
2nd Week
3rd Week
4th Week
Fenestra LC Dosing Schedule

All the tested mice including Balb/c, C57Bl/6, FVB and SCID groups underwent the repeat dosing schedule of Fenestra LC and VC as often as twice a week (10 mL/kg) or once a week (15 mL/kg) exhibited normal behavioral patterns and weight profiles throughout the study.

Conclusions

Acceptable injection tolerance and in vivo CT efficacious imaging profiles observed with both Fenestra LC and VC in single and multiple dosing regimes provide substantial flexibility and convenience for both anatomical and functional evaluations of the liver and vascular system in the same animal using microCT.

References

[1] Weber SM, et. al, J Surgical Research (2004) 119:41-45.
[2] Weichert JP, et. al, Radiology, (2000) 216:865-871.
[3] Weichert JP, et al., Academic Radiol (1998) 5:S16-19.
[4] Weichert JP, et. al, J Med Chem (1995) 38:636-646.

Supported in part by the UWCCC, UW-Radiology and Alerion Biomedical Inc.

gn.ton@hosp.wisc.edu, Poster #243